INFLUENCE OF A BUFFERED MATRIX AND PROTEIN UPON ALLICIN BIOAVAILABILITY FROM A COMMERCIAL GARLIC POWDER

R. Falkenberg and H. Archibald, Nutra Products, Inc., Fairfield, CA (Introduced by Doug Kalman, MS, RD, FACN)

Abstract

The ability of garlic powders to produce significant amounts of allicin upon disintegration in the stomach is unknown. We have assessed allicin bioavailability after garlic ingestion using breath analysis of allyl methyl sulfide (AMS), the primary allicin metabolite. We compared the allicin bioavailability of a buffered garlic product (BGz; Garli-Eze®, Nutra Products, Inc., Fairfield, CA) to a non-buffered form (NBG), both delivered in non-enteric coated capsules containing 600 mg of garlic powder. Two subjects were fed a high protein meal (HP) with water. Fifteen minutes after they began the meal they were given 3 capsules of BGz. The impact of a low protein meal (LP) prior to ingesting BGz or NBG capsules was also evaluated. Breath was collected before capsule consumption and an additional 18 times over the next 32 h. AMS was analyzed and expressed as area under the curve (AUC32 in ng-h/L). Percent allicin bioavailability (%AB) was determined by comparison to the AUC_{32} for consuming a known amount of allicin from crushed raw garlic. Results are expressed as the mean of the two subjects: AUC32: HP + BGz (1912), LP + BGz (2085), LP + NBG (1162); %AB: HP + BGz (85%), LP + BGz (93%), LP + NBG (51%). These data suggest that a buffered matrix augments the delivery and/or bioavailability of allicin, which also appears to operate independent of the buffering load of a protein-rich meal.

Rationale

Garlic has been the subject of intense clinical research, especially in relation to its hypolipidemic effects. The existing evidence base yields equivocal data, which likely is due to both substantial differences in study design and in the variable chemoprofiles of each of the garlic preparations that have been examined. This clinical evidence eclipses the number of clinical *pharmacologic* investigations performed to date. This may result from both the *a priori* assumption that any garlic preparation that yields garlic's primary bioactive, allicin, within an *in vitro* system would do so *in vivo*, and the absence of sufficiently specific and sensitive analytical methodologies. Allicin is a biologically ephemeral molecule that is produced only upon the crushing of garlic via the enzyme alliinase acting upon its substrate, alliin. Within dietary supplements, however, alliinase does not convert alliin to allicin until the dose form disintegrates in the body, requiring the preservation of alliinase activity throughout the manufacturing process as well as from the acidic and hydrolytic conditions accompanying gut transit. We have developed a novel composition that combines a natural alkalizing matrix with a suitably processed garlic extract (Garli-Eze®), designed to create a gut microenvironment fostering *de novo* allicin production and bioavailability, and obviating the need for enteric coating agents. We implemented a validated *in vivo* method shown to assess allicin bioavailability by capturing the breath evolution of the allicin end metabolite, Allyl Methyl Sulfide (AMS) (1).

Experimental Design

Subjects

- Two persons for whom 100% allicin bioavailability response had previously been determined after consumption of homogenized fresh garlic containing known amounts of allyl thiosulfinates (allicin and other thiosulfinates).

- Subject 1 (EM) = female, age 31, Ht. 5'5", Wt. 145 lbs
- Subject 2 (LL) = male, age 59, Ht. 5'6'', Wt. 140 lbs

Supplementation/Meal Protocol

- Subject restrictions: 24 hours prior to dosing - no garlic or onions or foods that commonly contain them in small amounts, such as salsa, ketchup, soups, salad dressings.

- The subjects ate 2 different standardized breakfast meals (8 am) on separate dates (min. 3 day washout):
 - High protein meal (27 g protein): a tuna sandwich (½ can, 3 oz., albacore tuna, 2 slices whole wheat bread, fat-free Miracle Whip) and 4 oz. of 2% milk.
 - Low protein meal (5 g protein): 2 slices of white bread toast (w/butter and jam), a banana, and 6 oz. water.
- At 15 min after starting to eat the meal (about 5-7 min after finishing the meal), each subject consumed 3 Garli-Eze capsules (2,200 mg garlic extract) or 3 capsules of a non-alkalized/non-buffered version of the same garlic extract (1,800 mg of garlic extract), with water (4-6 oz.).

Methods & Procedures

Allyl thiosulfinate potential analysis of garlic extract

- This value was necessary in order to determine % allicin bioavailability. This is the maximum amount of allicin and other allyl thiosulfinates that can be formed; specifically when the capsule contents are dispersed in aqueous solution, provided the final pH is in the optimum range for alliinase activity (pH 4.5-7). For non-buffered garlic powder, dispersion in water gave a pH in the optimum range. For Garli-Eze, it was necessary to disperse the powder in 0.015 N HCl, as dispersion in water gave a final pH of 8.7, which caused a considerable reduction in thiosulfinates. Allicin and other thiosulfinates were analyzed using C18-HPLC upon elution with methanol/water (1:1) at 240 nm (2).

Breath collection and analysis

- Subjects exhaled into 1-liter breath bags just before consuming the meal and capsules, then hourly for 8 hours, then every two hours (except during sleep) until 32 hours after capsule consumption. Each subject was given 4 breath bags to take home for breaths at 10, 12, 14, and 22 hours after consumption. Non-leak bags were used for overnight breaths.

- Breath samples were analyzed for AMS using a sensitive, sulfur-selective detector (GC-PFPD) (1). The area under the 32-hour concentration curve (AUC_{32}) was determined by a computer program. Bioavailability, relative to consumption of a known amount of allicin from raw, crushed garlic, was expressed as a percentage.

Data Analysis

- Data are presented as the changes from baseline for each subject, LL and EM, or as the mean of these 2 subjects.

Results

- The protein content of the meal appeared to have no impact upon mean allicin bioavailability of Garli-Eze, being 85.0% for the high protein meal and 92.5% for the low protein meal (***top right image***).
- Under the low protein meal conditions the mean allicin bioavailability of the non-alkalized/non-buffered garlic composition was 51.0% (***middle right image***).
- Neither subject reported gastrointestinal discomfort or other adverse effects in association with the ingestion of either garlic composition.

Conclusions

- Under the conditions employed in this pilot, proof of concept study, the Garli-Eze alkalized/buffered garlic composition demonstrated bioavailability comparable to a similar amount of allicin released from raw, crushed garlic (***bottom right image***).
- The protein content of the test meals appeared to have negligible impact upon allicin bioavailability.
- Allicin bioavailability of the non-alkalized garlic composition, which lacked enteric coating, was unexpectedly high.
- Further investigations examining the pharmacokinetics and pharmacodynamics of Garli-Eze are warranted.

References

1. Lawson LD and Wang ZJ. Allicin and allicin-derived garlic compounds increase breath acetone through allyl methyl sulfide: use in measuring allicin bioavailability. *J Agric Food Chem* 2005;53:1974-83.
2. Lawson LD, Wood SG, Hughes BG. HPLC analysis of allicin and other thiosulfinates in garlic clove homogenates. *Planta Med* **1991**;*57*:263-70.

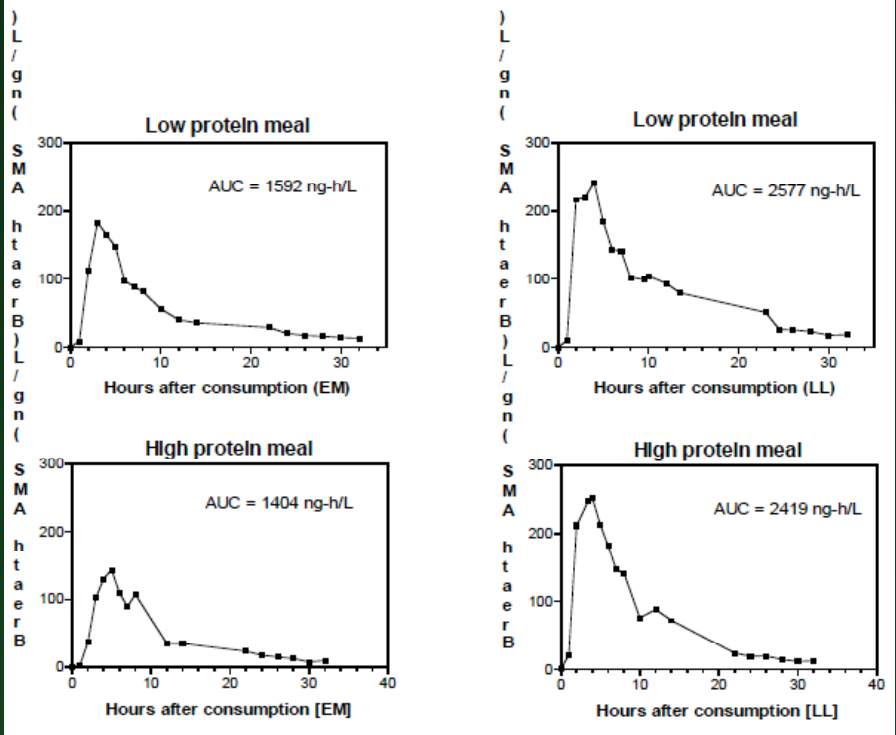
Low protein meal
AUC = 1592 ng-h/L
Hours after consumption (EM)
Low protein meal
AUC = 2577 ng-h/L
Hours after consumption (LL)
High protein meal
AUC = 1404 ng-h/L
Hours after consumption [EM]
High protein meal
AUC = 2419 ng-h/L
Hours after consumption [LL]
Breath AMS (ng/L)

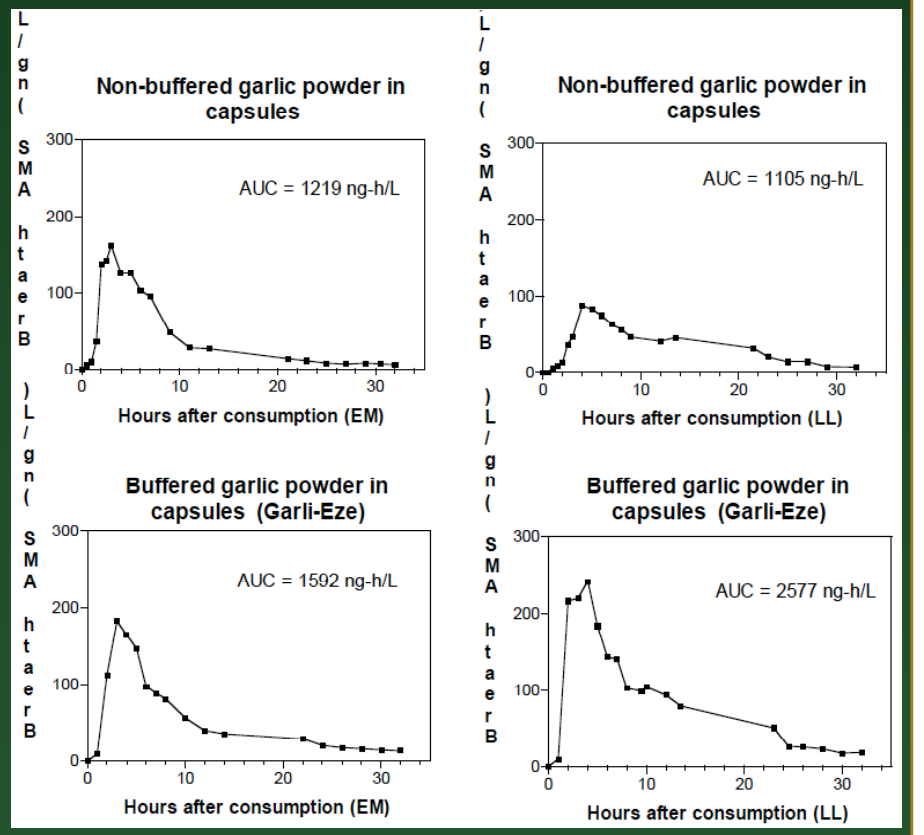
Non-buffered garlic powder in capsules
AUC = 1219 ng-h/L
Hours after consumption (EM)
Non-buffered garlic powder in capsules
AUC = 1105 ng-h/L
Hours after consumption (LL)
Buffered garlic powder in capsules (Garli-Eze)
AUC = 1592 ng-h/L
Hours after consumption (EM)
Buffered garlic powder in capsules (Garli-Eze)
AUC = 2577 ng-h/L
Hours after consumption (LL)
Breath AMS (ng/L)

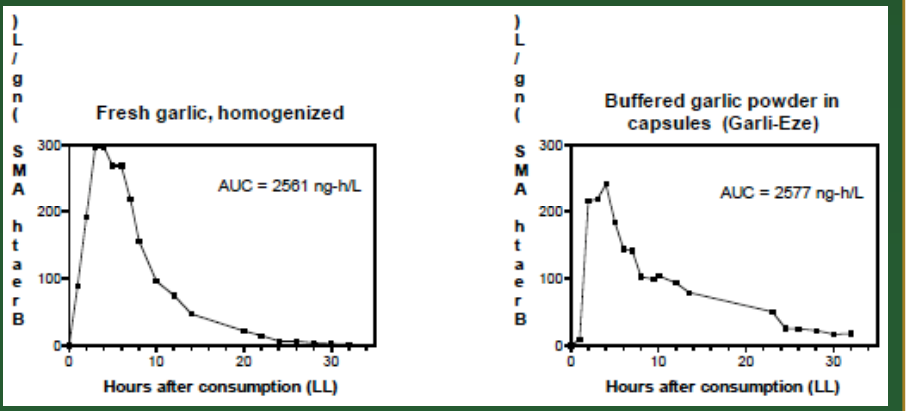
Fresh garlic, homogenized
AUC = 2561 ng-h/L
Hours after consumption (LL)
Buffered garlic powder in capsules (Garli-Eze)
AUC = 2577 ng-h/L
Hours after consumption (LL)
Breath AMS (ng/L)